RESTORE

Reassessing Psychotropic Burden, Functional Recovery, and Stimulant Relapse After Incarceration

A hypothesis-generating clinical framework and proposed evaluation

Hughes Mohlie, PMHNP-BC

BACKGROUND

Community reentry requires alertness, motivation, emotional responsiveness, planning, concentration, and independent functioning. Some psychotropic regimens may add sedation or other treatment burden that can complicate these demands. Medication effects must be distinguished from illness-related symptoms and social stressors.

CLINICAL OBSERVATION & RESEARCH GAP

In post-incarceration care, some patients describe methamphetamine as temporarily restoring energy, focus, motivation, or emotional responsiveness - not only as producing euphoria. This observation does not establish prevalence, mechanism, or causation. The possible pathway from treatment burden to impaired function to stimulant use has not been established and warrants systematic study.

METHODS / EVALUATION (PROPOSED)

Prospective, mixed-methods feasibility cohort during community reentry.

1. Reconcile diagnoses, medications, doses, timing, and cumulative sedative/anticholinergic burden.
2. Repeatedly assess sleepiness, anhedonia/amotivation, cognition, psychiatric symptoms, daily functioning, stimulant craving/use, and reentry conditions.
3. Use structured interviews to characterize perceived motives and effects of methamphetamine.
4. Evaluate within-person change after clinically indicated treatment optimization.

Primary aim: feasibility and signal detection; not causal inference.

THE RESTORE MODEL

A sequence of testable questions - not an established causal pathway

1 — PSYCHOTROPIC BURDEN → FUNCTION

Is greater post-release psychotropic or sedative burden associated with impaired functional recovery?

2 — FUNCTIONAL IMPAIRMENT → METHAMPHETAMINE USE

For some individuals, is methamphetamine use partly an attempt to restore perceived functioning?

3 — TREATMENT OPTIMIZATION → FUNCTIONAL RECOVERY

Can systematic reassessment and individualized optimization improve real-world functioning?

4 — FUNCTIONAL RECOVERY → RELAPSE

If functioning improves, is stimulant relapse or continued use reduced?

CORE HYPOTHESIS TO BE TESTED
Optimizing indicated psychiatric treatment may support function - and may reduce one underrecognized driver of stimulant use.

Alternative explanations and confounding will be assessed; appropriate treatment is never withdrawn solely for research.

PROGRESS TO DATE / PRELIMINARY OBSERVATIONS

The clinical observation has been translated into a four-question logic model, candidate measurement domains, and a proposed longitudinal evaluation. No participant outcome data are reported here. The framework remains in development and is intended to generate testable hypotheses.

DEI / HEALTH EQUITY

People returning from incarceration experience disproportionate psychiatric, substance-use, housing, employment, and care-access barriers. Evaluation should use person-first language, include patient-reported function and goals, monitor differential burden by race/ethnicity, gender, disability, and housing status, and avoid coercive or stigmatizing assumptions. Medication review must preserve access to indicated treatment.

CONCLUSION / IMPACT

RESTORE shifts the clinical lens from symptom control alone to symptom control plus functional recovery. The framework invites careful study of medication burden, patient-perceived stimulant effects, and relapse - without presuming that any link is causal.

CLINICAL PRACTICE TAKE-HOME MESSAGES

1. After release, reassess diagnoses, medication burden, sedation, cognition, and daily function - while maintaining necessary treatment.

2. Ask what methamphetamine is perceived to do for the patient; the answer may reveal functional treatment targets without minimizing addiction risk.

REFERENCES

1. Reeve S, Robbins K, Hodgekins J. Psychiatry Res. 2025;351:116641. doi:10.1016/j.psychres.2025.116641.
2. Huhn M, et al. JAMA Psychiatry. 2024;81(10):1006-1018. doi:10.1001/jamapsychiatry.2024.2063.
3. Sekhon S, et al. BMC Health Serv Res. 2022;22:1300. doi:10.1186/s12913-022-08656-7.
4. Hart CL, et al. Psychopharmacology. 2001;157:75-81. doi:10.1007/s002130100738.

RESTORE webpage

EXPLORE THE RESTORE PROJECT

Scan for the evolving framework, evidence, references, and updates.

Research & Collaboration

hughes@moodclinics.com

moodclinics.com/restore

IN PROGRESS • HYPOTHESIS-GENERATING • NO CAUSAL CLAIMS OR PARTICIPANT OUTCOMES